A Dynamic Light Scattering (DLS) Assay for the Analysis of Aggregated Adenovector Particles

Jonathan K. Smith, Marilyn Menger, Miguel Carrion and Bryan Butman

GenVec, Inc., Gaithersburg, MD 20878

Introduction

The complex process by which viral vectors are manufactured, formulated and stored can provide numerous opportunities for viral protein modifications that can possibly lead to vector particle aggregation. Conventional biochemical methods for analysis of the viral proteome (SDS-PAGE, reversed-phase HPLC, MALDI-TOF) are disruptive to the structure and organization of the viral particles and therefore do not provide sufficient insight into the aggregation phenomenon. Development of dynamic light scattering (DLS) methods has enabled analysis of large proteomes in their native form. The objective of this work is to demonstrate the potential for use of DLS for monitoring and quantitating the presence of aggregates among Adenovectors whose components may have undergone a physical change.

The following criteria were considered to be essential in the development of a light scattering method for Adenovector preparations: Analysis of vectors in their native state, determination of the mean particle size in the vector population, characterization of the homogeneity (polydispersity index and graphical profile) within a vector sample, and suitability of the assay for the QC lab environment (Ease of use, robust performance, and reproducibility). The Malvern HPPS 5001automated particle sizer meets these design criteria.

System Suitability – Latex Size Standards

60 nm Latex Std., Z-Ave. = 63.3 nm , PDI = 0.031

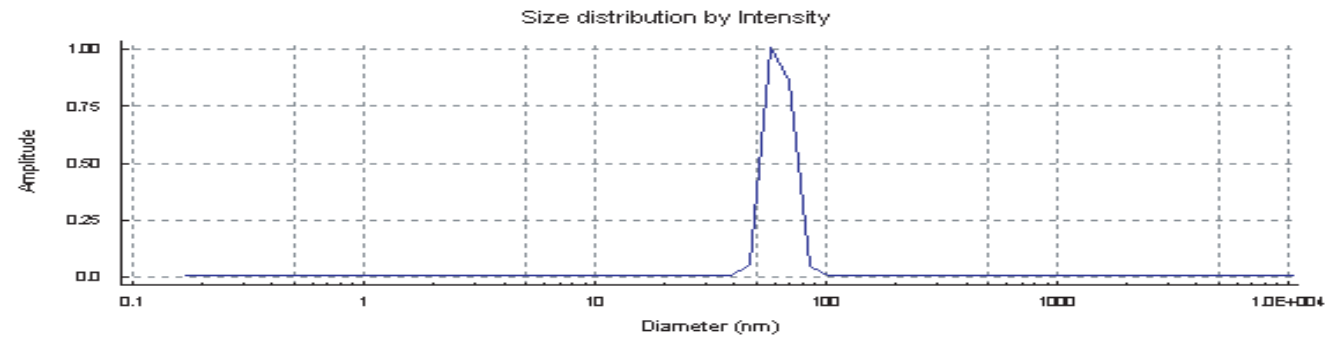

Size distribution by Intensity
Amplitude
Diameter (nm)

90 nm Latex Std., Z-Ave. = 92.1 nm, PDI = 0.016

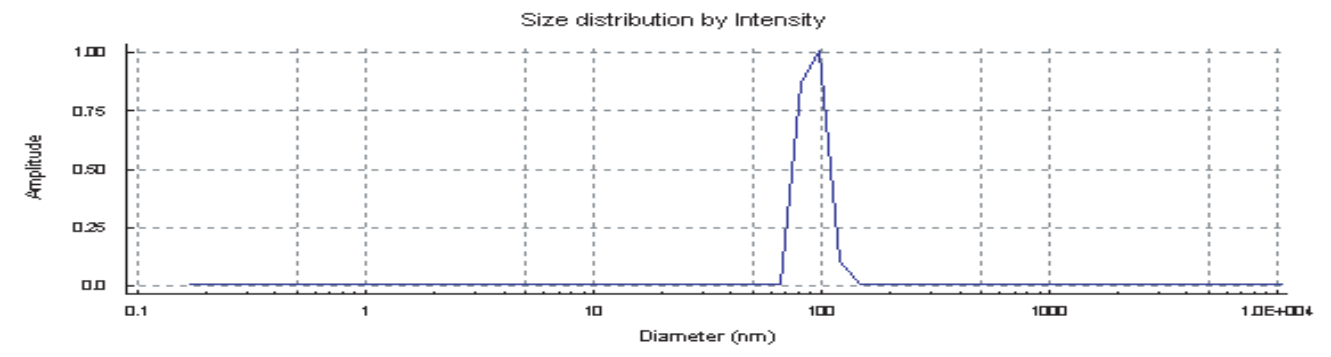

Size distribution by Intensity
Amplitude
Diameter (nm)

200 nm Latex Std., Z-Ave. = 198.1nm, PDI = 0.016

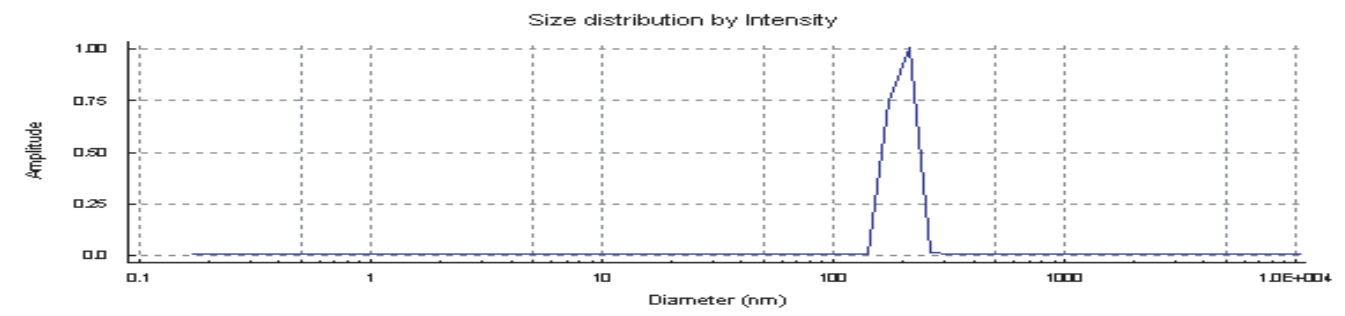

Size distribution by Intensity
Amplitude
Diameter (nm)

	System Suitability - Distribution by Intensity / Volume				
Size Standard	Polydispersity Index [Avg.]	Acceptable Polydispersity Index	Z-Average Size (nm) [Avg. of Runs]	Acceptable Z-Avg. Size Range (nm)	Peak Size (Mode)
60 nm Latex Standard	0.02 +/- 0.01	< 0.2	63.00 +/- 0.20	61.04 - 65.56	62.87 +/- 0.38
90 nm Latex Standard	0.01 +/- 0.01	< 0.2	90.75 +/- 0.30	87.48 - 91.56	91.08 +/- 0.29
200 nm Latex Standard	0.03 +/- 0.01	< 0.2	198.04 +/- 0.99	194.84 - 199.96	196.57 +/- 0.82

Figure 1. Conditions established for the analysis of the NIST certified latex standard at 25°C in a quartz cuvette. The bracketing of the size range provides a suitable level of sensitivity and accuracy.

Prior to analyzing any test articles, the Adenovector's sample matrix (viscosity and refractive index) is taken into consideration in the instrument software. In Figure 2, an ideal spectrum is presented for a purified vector preparaton, illustrating a homogenous peak (unaggregated) with a polydispersity index (PDI) less than 0.2 and a Z-Ave within 90 – 110 nm. This data meet the established assay acceptance criteria.

Native Vector Analyzed via DLS

Z-Ave. = 104.9 nm, PDI = 0.037

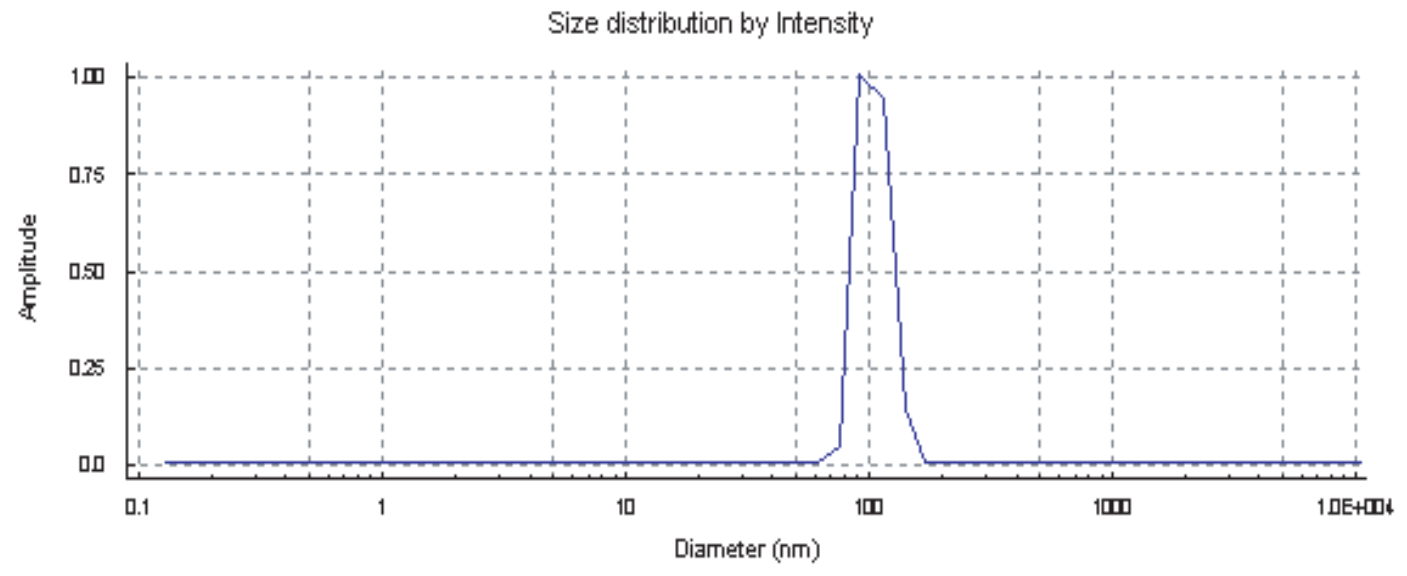

Size distribution by Intensity
Amplitude
Diameter (nm)

Figure 2. Illustration of a suitable Adenovector Reference Standard analyzed using an established system SOP method for the presence of aggregates. The Malvern HPPS 5001 system provides a value of 104.9 nm (Z-Ave.) , which falls within the theoretical size of an Adenovector reported between 90 – 105 nm. The Ref. Std. also has a low PDI value (0.037).

Aggregation Analysis via DLS (Protein/Ab Interaction)

Native,
PDI=0.052
104.5mm

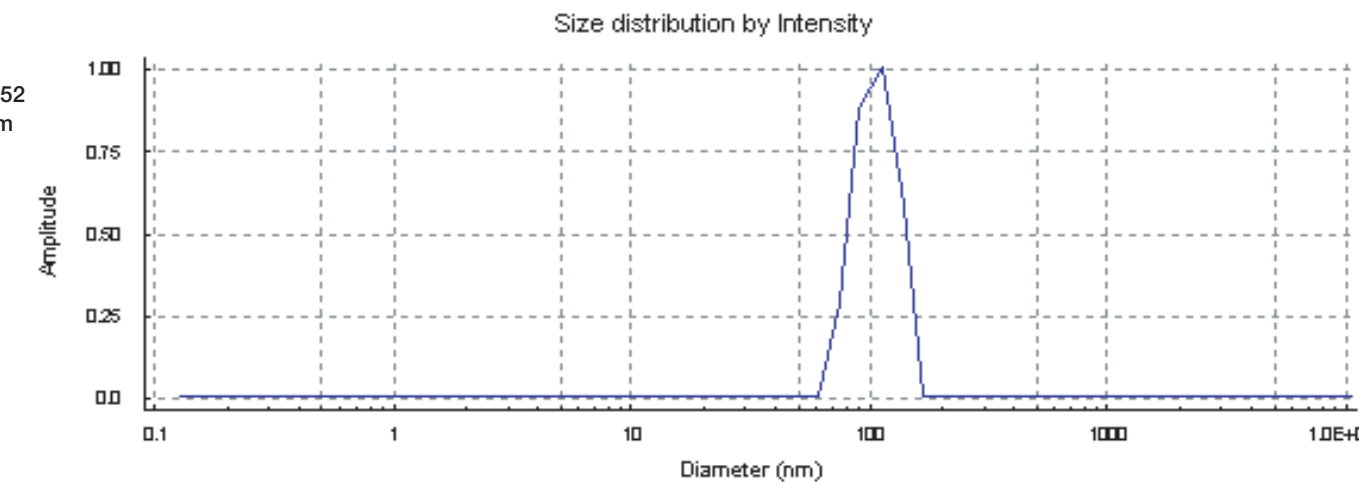

Size distribution by Intensity
Amplitude
Diameter (nm)

1:100w/1°Ab
PDI=0.542
2927.9 mm

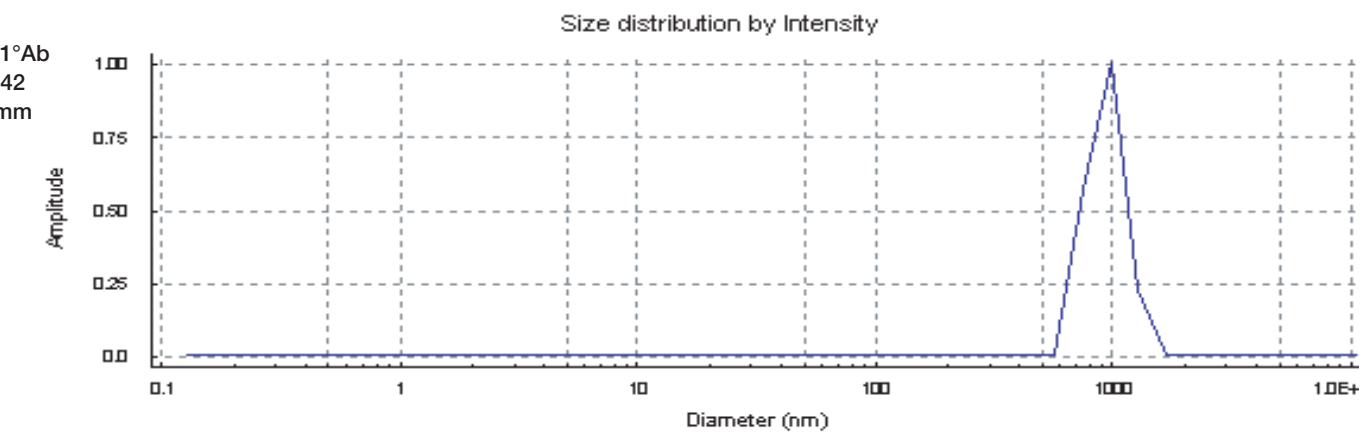

Size distribution by Intensity
Amplitude
Diameter (nm)

Figure 3. A model system for vector aggregation involves the use of polyclonal antibodies against Ad5 WT total proteins. The first spectrum presented above provides an initial view of the sample prior to the addition of a polyclonal antibody (1:100). In the second spectrum, the sample was allowed to incubate at room temperature for 30 minutes and then equilibrated in the system for 5 minutes at 25°C prior to analyzing. The size of the native sample completely shifted, indicating the formation of large clusters (aggregates).

Time = Day 0, Z-Ave. = 93.1 nm, PDI = 0.006, Peak % = 100

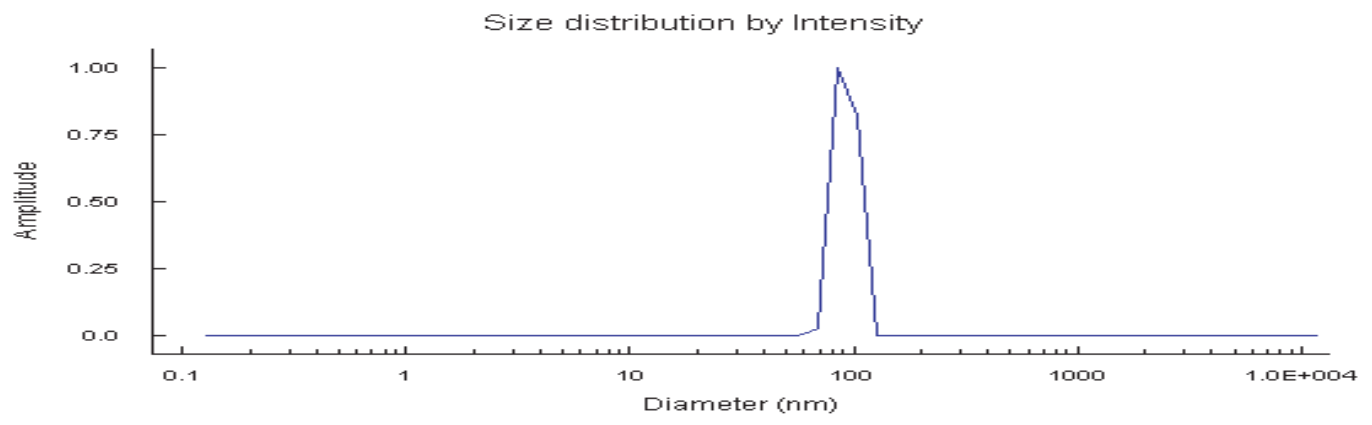

Size distribution by Intensity
Amplitude
Diameter (nm)

Time = Day 7, Z-Ave. = 115.6 nm, PDI = 0.369, Peak % = 72.4

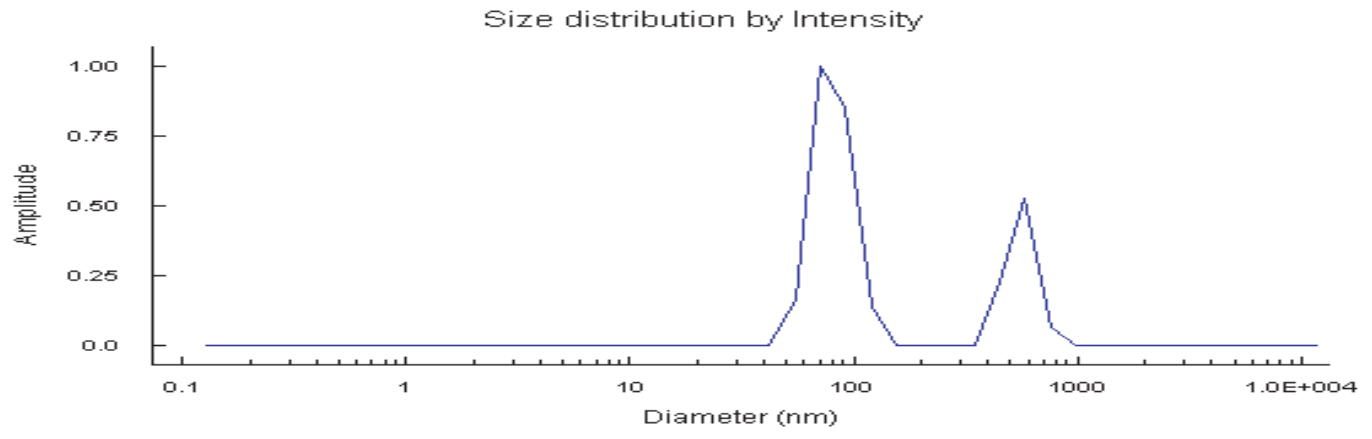

Size distribution by Intensity
Amplitude
Diameter (nm)

Time = Day 14, Z-Ave. = 171.0 nm, PDI = 0.473, Peak % = 58.2

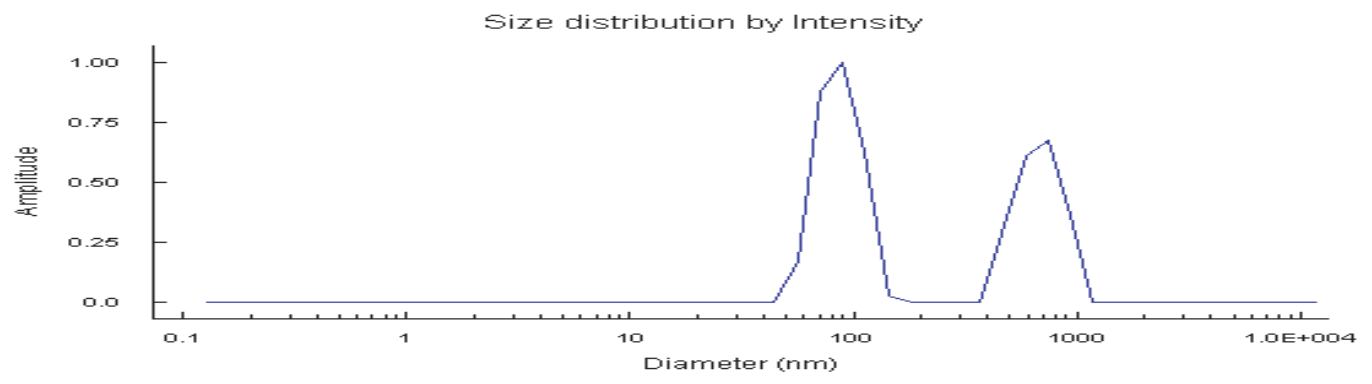

Size distribution by Intensity
Amplitude
Diameter (nm)

Time = Day 21, Z-Ave. = 864.5 nm, PDI = 0.475, Peak % = 16.2

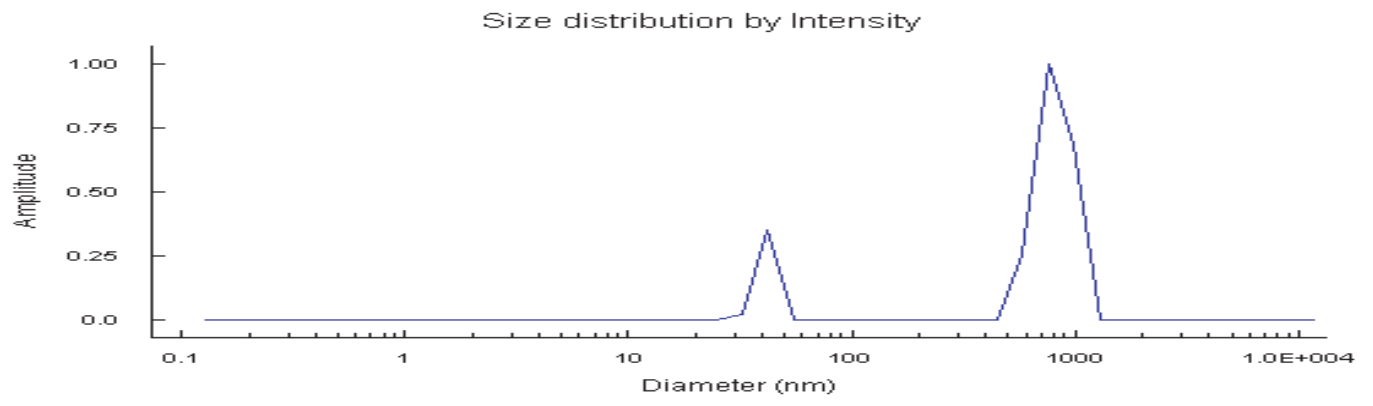

Size distribution by Intensity
Amplitude
Diameter (nm)

Figure 4. Vector particle aggregates can also be generated by incubating a purified Adenovector preparation at 37°C. By Day 7, a second peak is detected at approximately 600 nm and the sample PDI has increased to >0.300. By Day 21, most of the vector has formed aggregates, with a Z_{ave} value of >800 nm and PDI >0.400

Accelerated Stability Analysis - 37C, Intensity Distribution

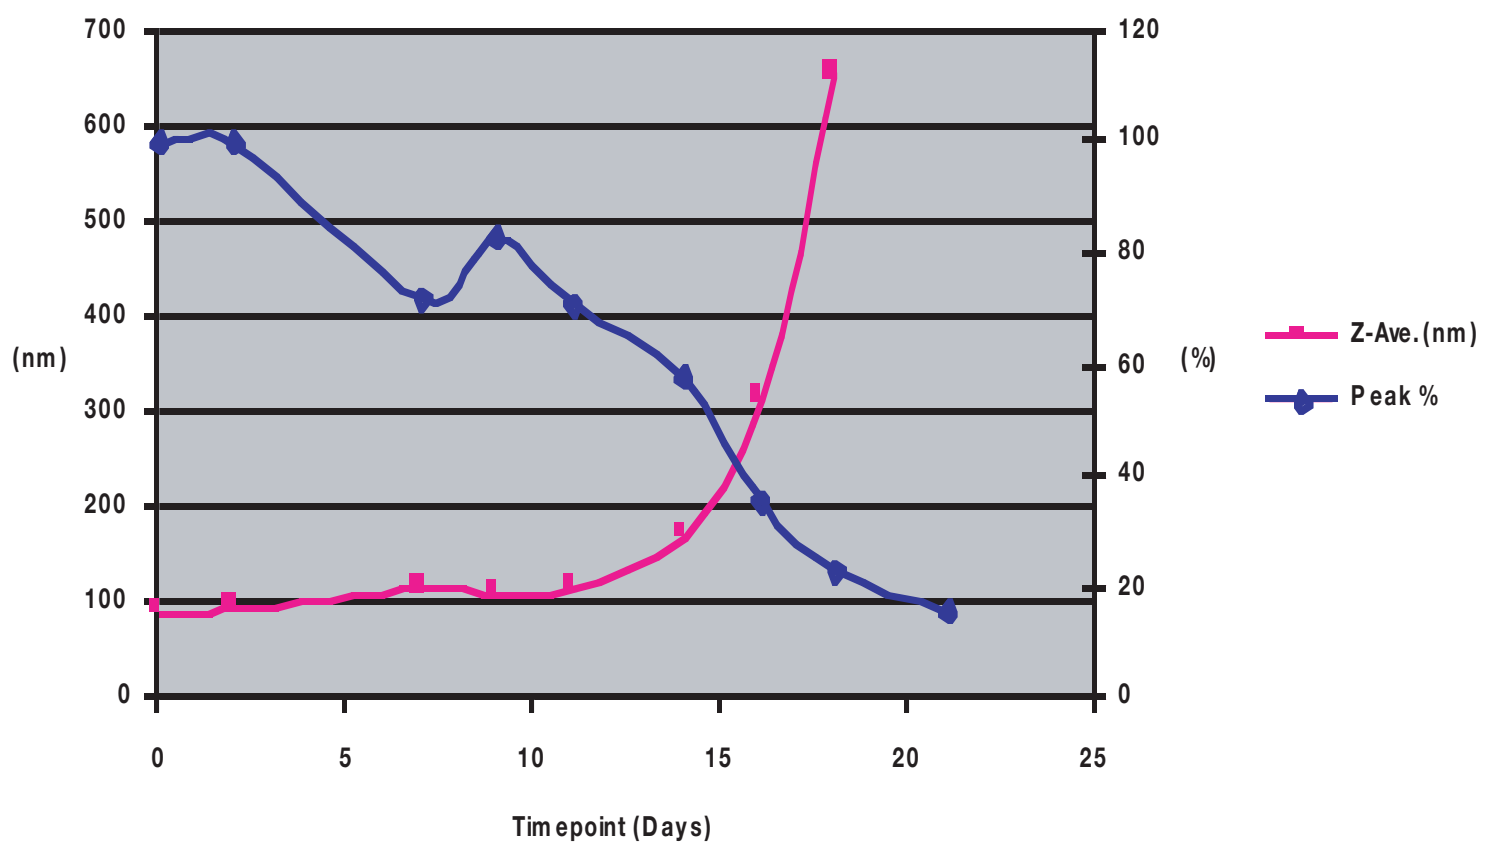

(nm)
(%)
Z-Ave. (nm)
Peak %
Timepoint (Days)

Intensity Distribution

Time Point	Monomeric Peak %	PDI_
0 Days	100	0.006
7 Days	72	0.369
14 Days	58	0.473
21 Days	16	0.476

Figure 5. Graphical representation of the loss of monomeric vector peak % and the increase in the PDI as a function of time at 37°C.

Summary:

- An aggregation assay has been developed for Adenovirus vectors based upon dynamic light scattering (DLS) using the Malvern Instruments HPPS 5000 instrument
- Instrument software generates an "SOP" procedure that enables reproducible sample analysis and the generation of a formal report
- System suitability parameters have been defined using mono-disperse latex spheres and an Adenovector Reference Standard
- The Malvern instrument generates the following analytical data on test articles:
 - Visual peak profile
 - Mean Particle Size (Z-Ave) and Polydispersity Index values
 - Multiple peak analysis within a sample
 - Individual peak size, peak area percent (of total)
- This assay is useful for release testing and stability monitoring of purified Adenovector lots

GENVEC